Neoadjuvant pembrolizumab plus lenvatinib in resectable stage III melanoma patients (NeoPele); analysis of tumor microenvironment (TME) correlated to pathological response.

Ines Pires da Silva[1,2,3,4,5], Jordan W. Conway[1,2,3,4], Akshaya Ramanathan[1], Laura Smith[1], Robert Rawson[1,2,4,6], Piyush Grover[1], Nigel Maher[1,2,3,4], Jorja Braden[1,2,3,4], Maria Gonzalez[1], Umaimainthan Palendira[3,4], Alexander A. Menzies[1,4,7], Richard Scolyer[1,2,3,4,6], Georgina V. Long[1,2,3,4,7]

[1]Melanoma Institute Australia, Sydney, Australia; [2]Faculty of Medicine and Health, Sydney, Australia; [3]Charles Perkins Centre, Sydney, Australia; [4]The University of Sydney, Sydney, Australia; [5]Crown Princess Mary Cancer Centre Westmead, Blacktown Hospital, Sydney, Australia; [6]Royal Prince Alfred Hospital - Pathology Department, Sydney, Australia; [7]Royal North Shore and Mater Hospitals, Sydney, Australia.

Background

- ❑ The phase II SWOG S1801 study showed an improved event-free survival with anti-PD-1 (PD1) neoadjuvant immunotherapy (neoIT) *versus* adjuvant PD1[1].
- ❑ One hypothesis explaining this benefit is the presence of tumor-draining lymph nodes (tdLN; defined as the nearest node to the tumor without direct involvement) as a potential reserve of stem-like (TCF7+) T cells, crucial to a good response to immunotherapy[2,3].

Objectives

We sought to analyze the immune infiltrate of the tumor-involved LN (i.e. TME) and tdLN from patients achieving major pathological response (***MPR***: complete [pCR] or near-complete [near-pCR] pathological response) *versus* ***non-MPR*** (partial [pPR] or no [pNR] pathological response).

Methods

- ❑ Patients with clinical stage III melanoma treated with 6 weeks of PD1-based neoIT (PD1 + Lenvatinib) were included (NeoPele clinical trial; NCT04207086).
- ❑ Multiplex fluorescent immunohistochemistry of the TME before and after neoIT, and of the tdLN after neoIT was analyzed:
 - T cell panel: CD3, TCF7, CD103, FoxP3, CD39 and Sox10;
 - B cell panel: CD20, CD21, CXCR5, TCF7, CD3 and SOX10.
 - Lymphoid aggregates, characterized by clusters of CD21+ and CXCR5+ immune cells, were quantified.

Results

FIGURE 1. Study design[4].

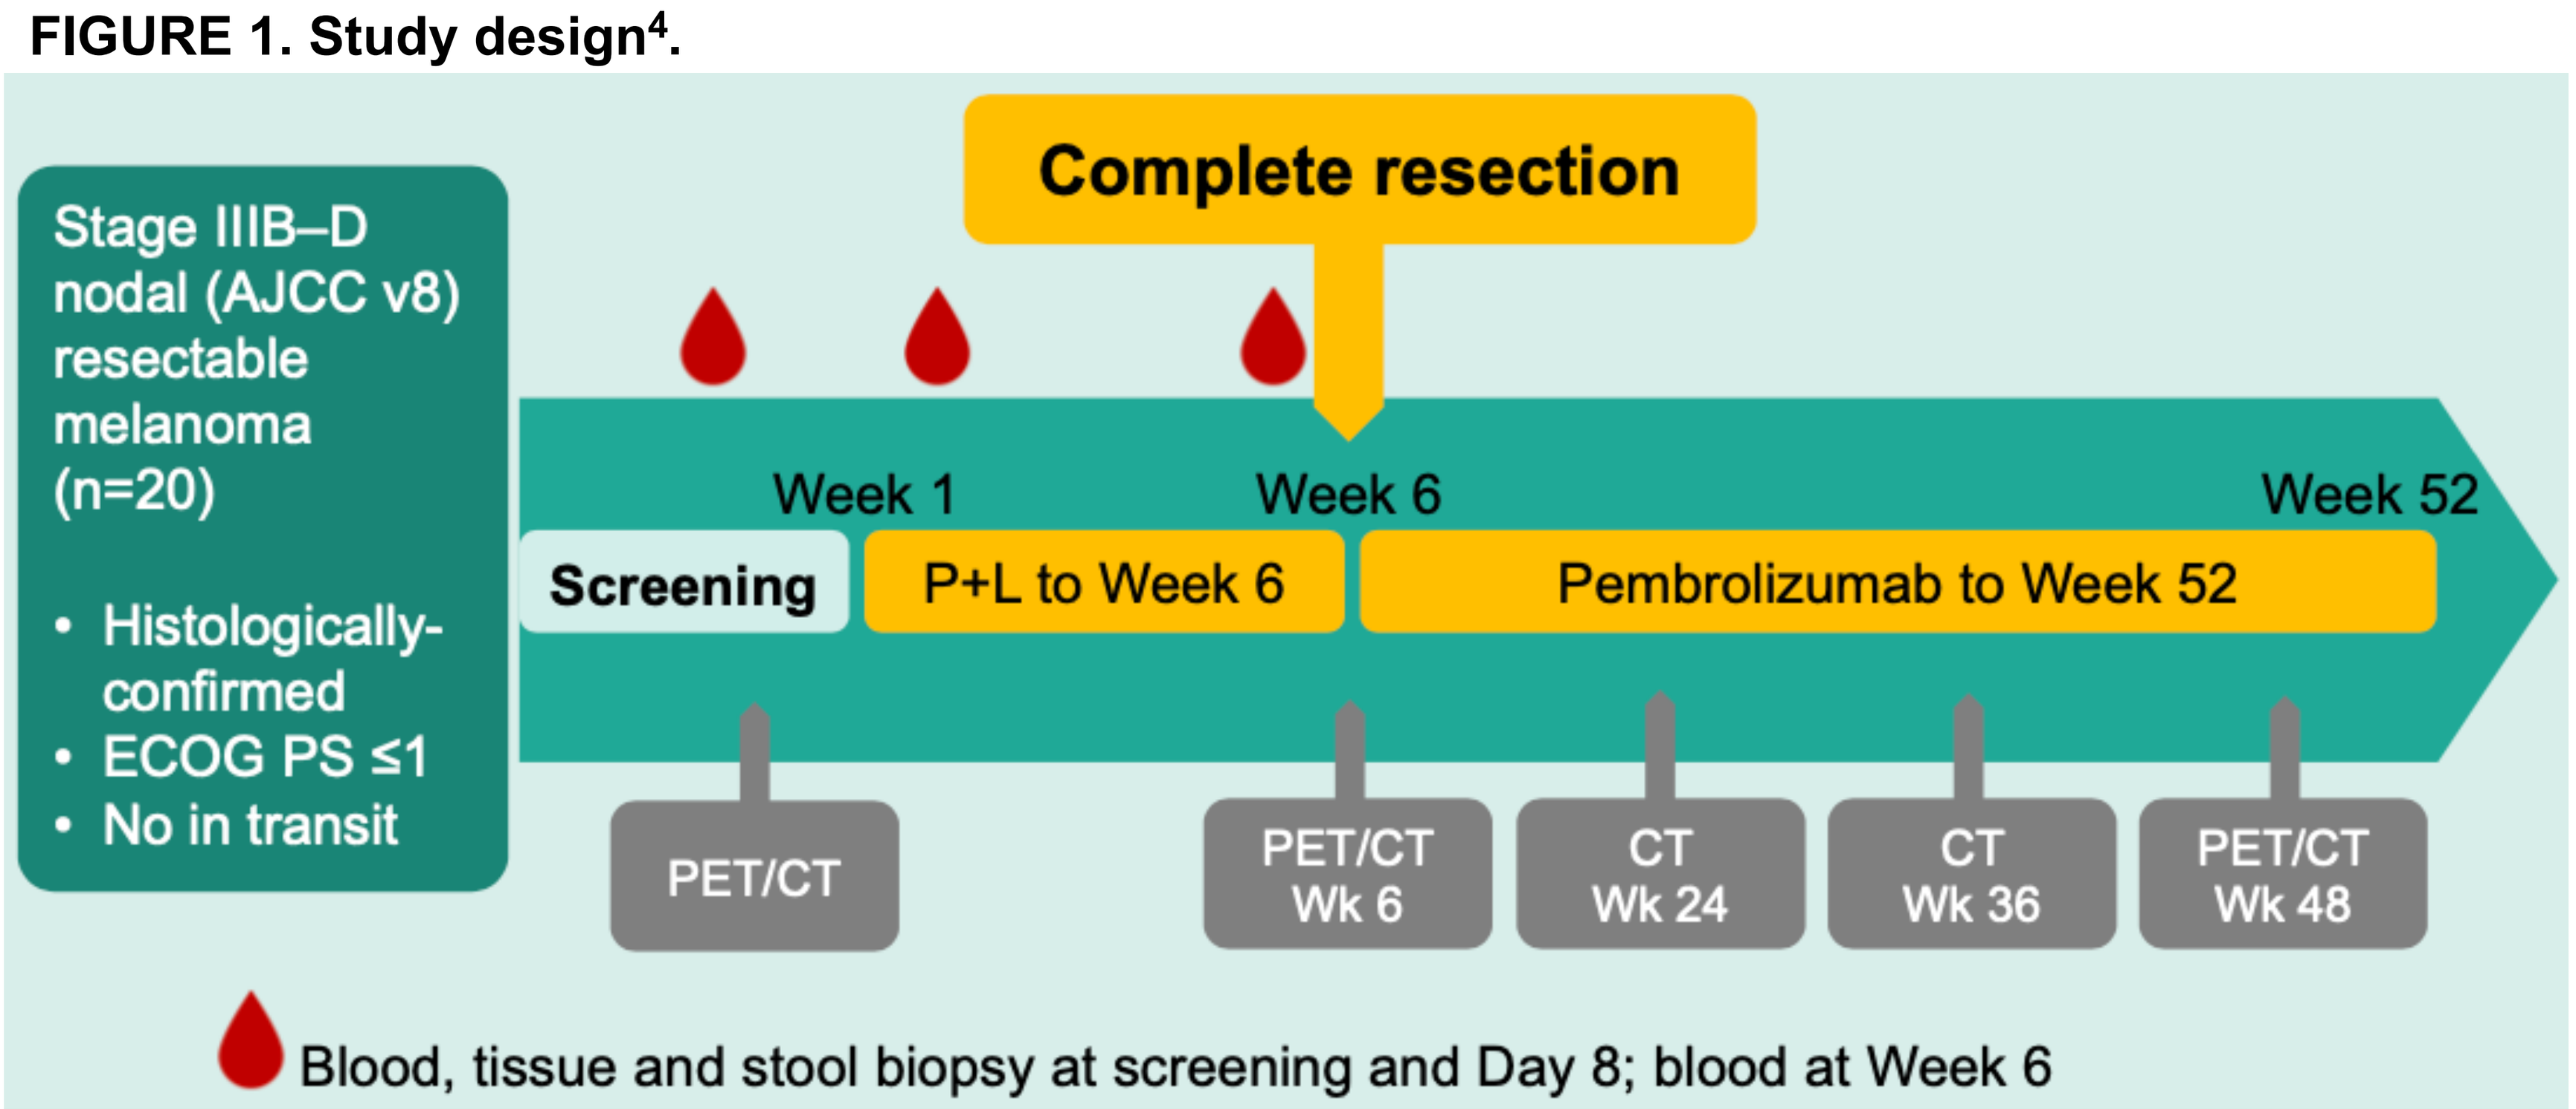
Complete resection
Stage IIIB–D nodal (AJCC v8) resectable melanoma (n=20)
• Histologically-confirmed
• ECOG PS ≤1
• No in transit
Week 1
Week 6
Week 52
Screening
P+L to Week 6
Pembrolizumab to Week 52
PET/CT
PET/CT Wk 6
CT Wk 24
CT Wk 36
PET/CT Wk 48
Blood, tissue and stool biopsy at screening and Day 8; blood at Week 6

Results: *MPR (n=11, 55%) versus non-MPR (n=9, 45%)*

1. Baseline

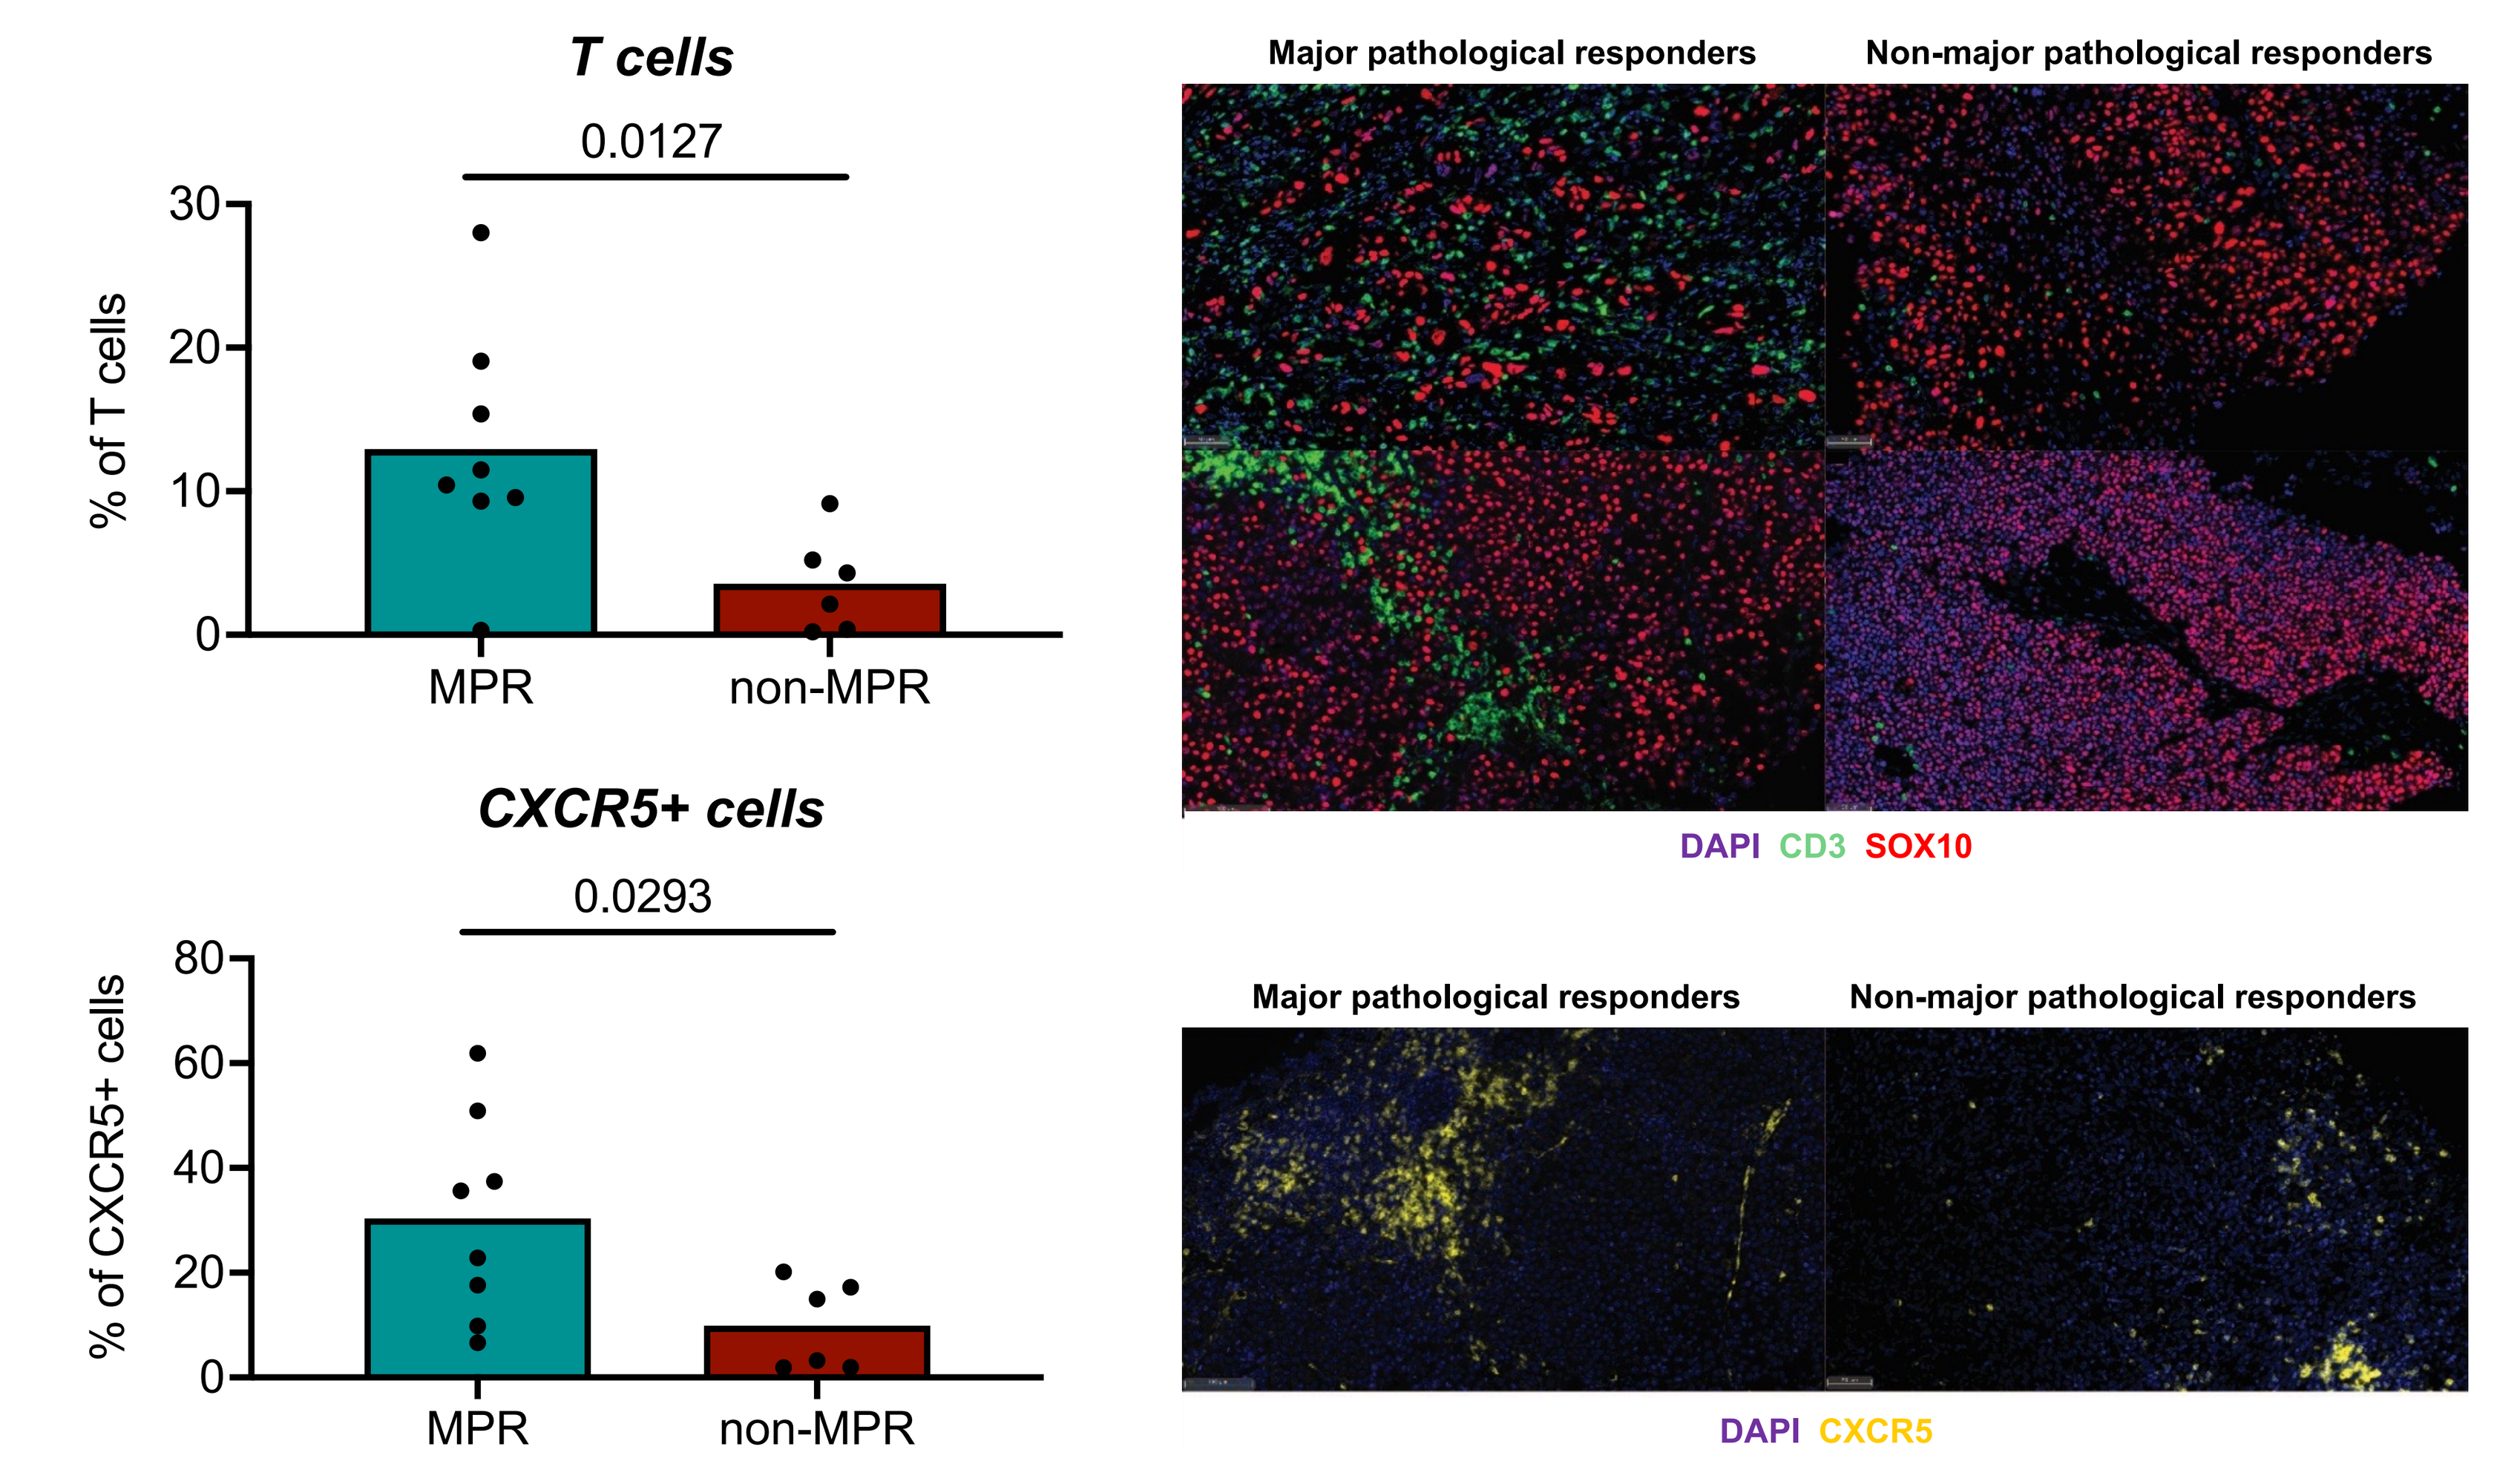
T cells
0.0127
% of T cells
MPR
non-MPR
Major pathological responders
Non-major pathological responders
DAPI CD3 SOX10
CXCR5+ cells
0.0293
% of CXCR5+ cells
MPR
non-MPR
Major pathological responders
Non-major pathological responders
DAPI CXCR5

FIGURE 2. Differences in the immune infiltrate of the TME between MPR *versus* non-MPR patients at baseline.

3. Week 6

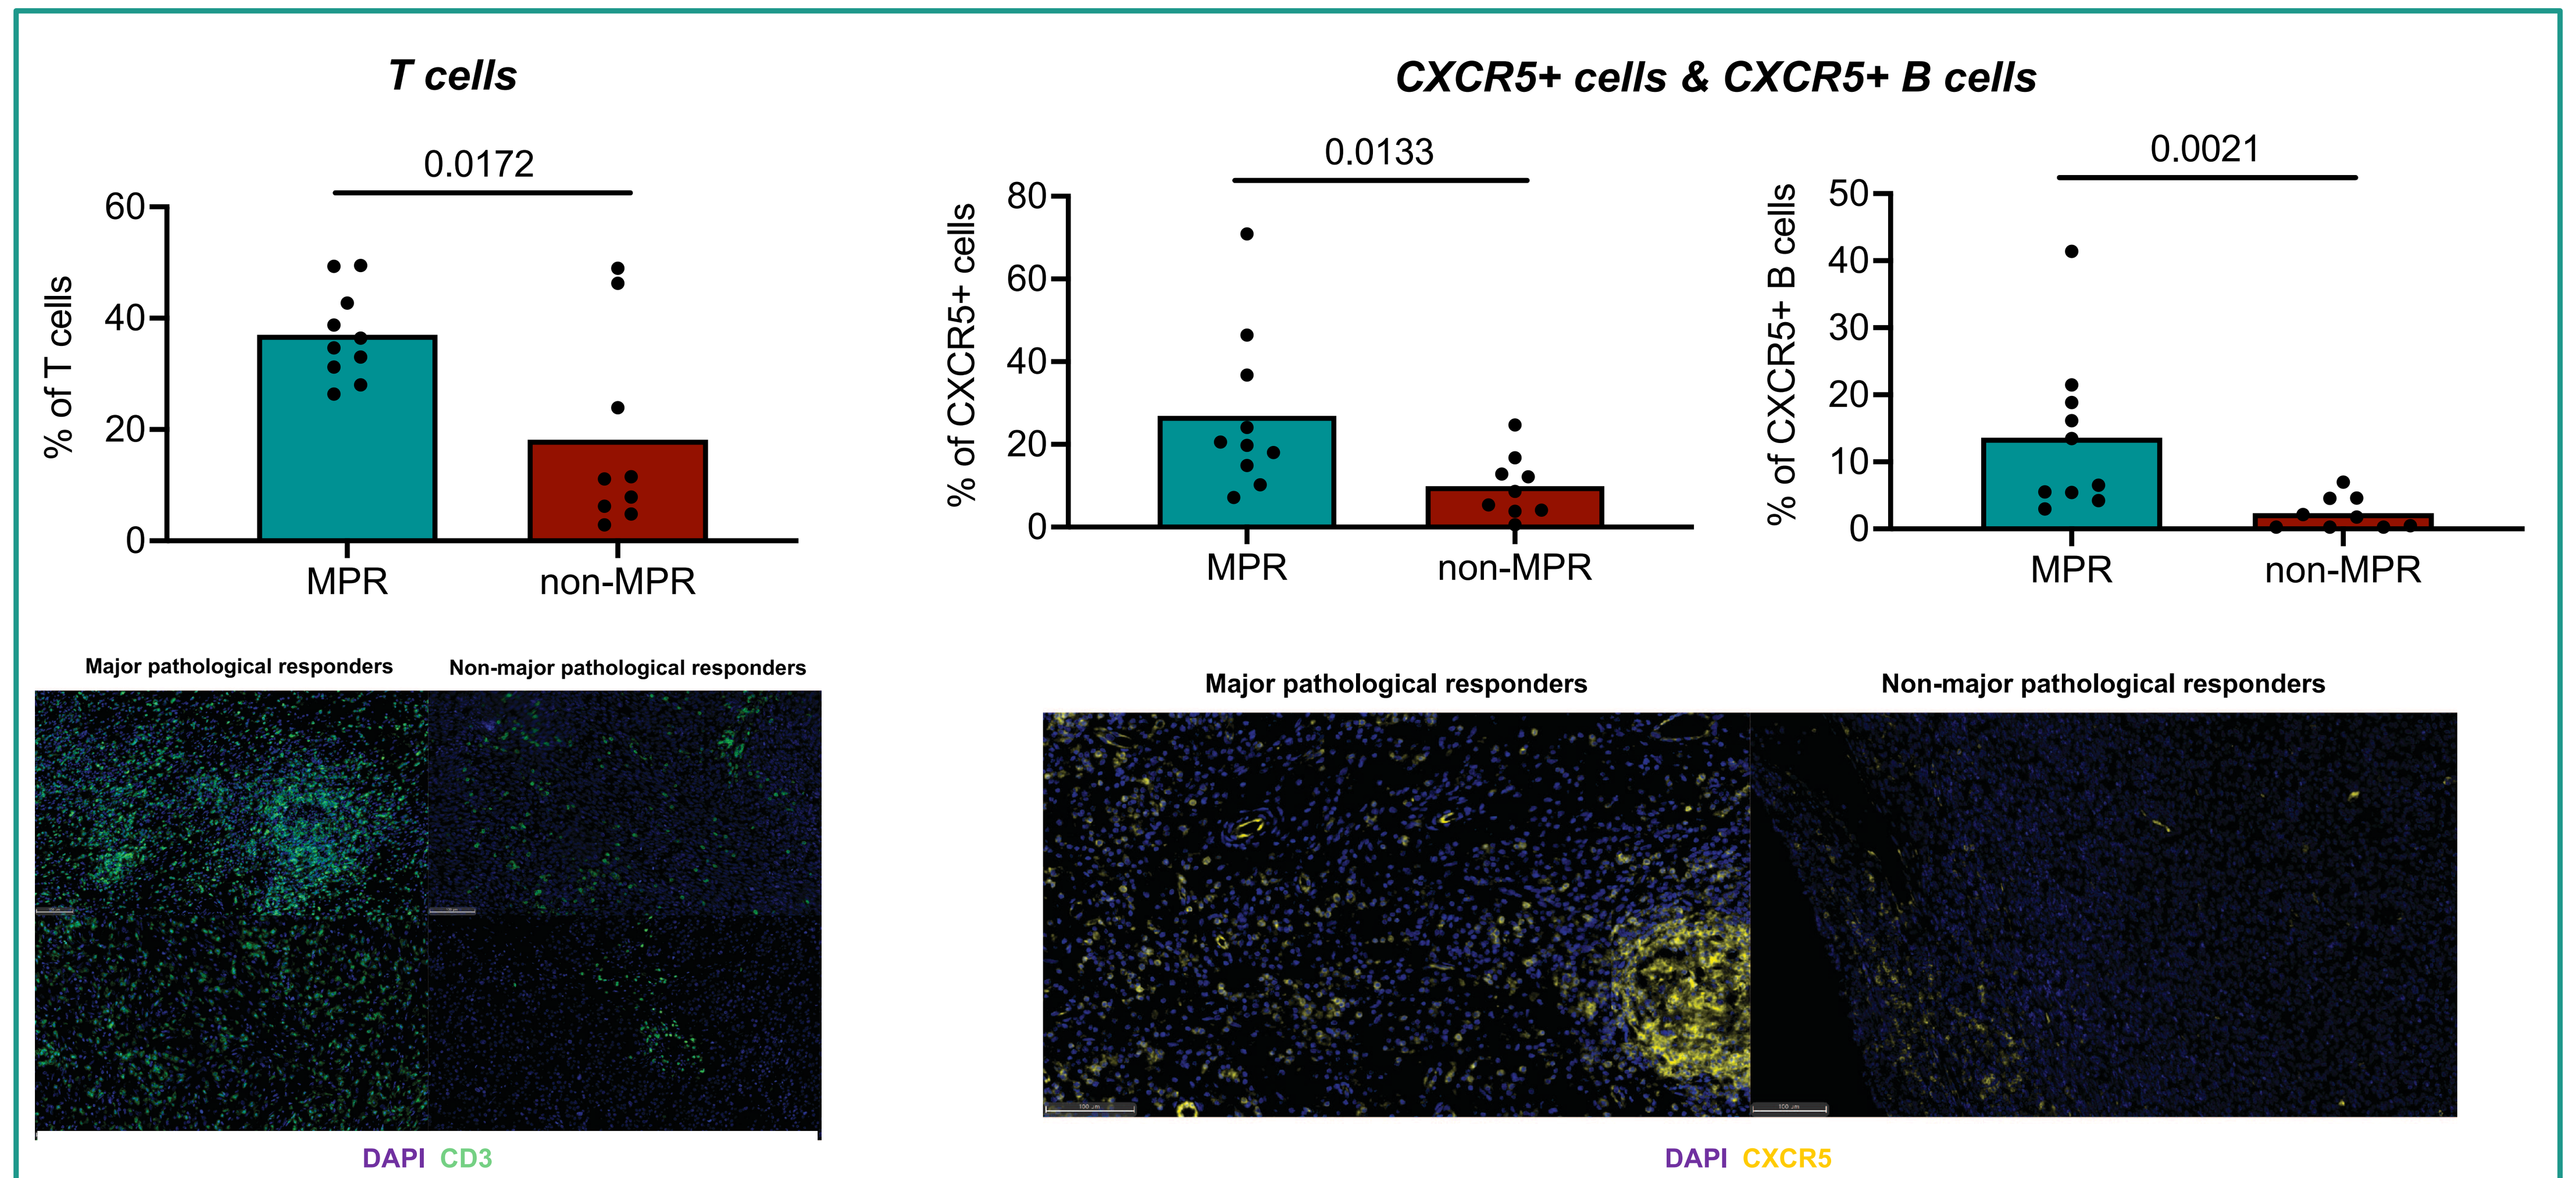
T cells
0.0172
% of T cells
MPR
non-MPR
CXCR5+ cells & CXCR5+ B cells
0.0133
% of CXCR5+ cells
0.0021
% of CXCR5+ B cells
Major pathological responders
Non-major pathological responders
DAPI CD3
DAPI CXCR5

FIGURE 4. Differences in the immune infiltrate of the TME between MPR *versus* non-MPR patients at week 6.

2. Baseline → Week 6

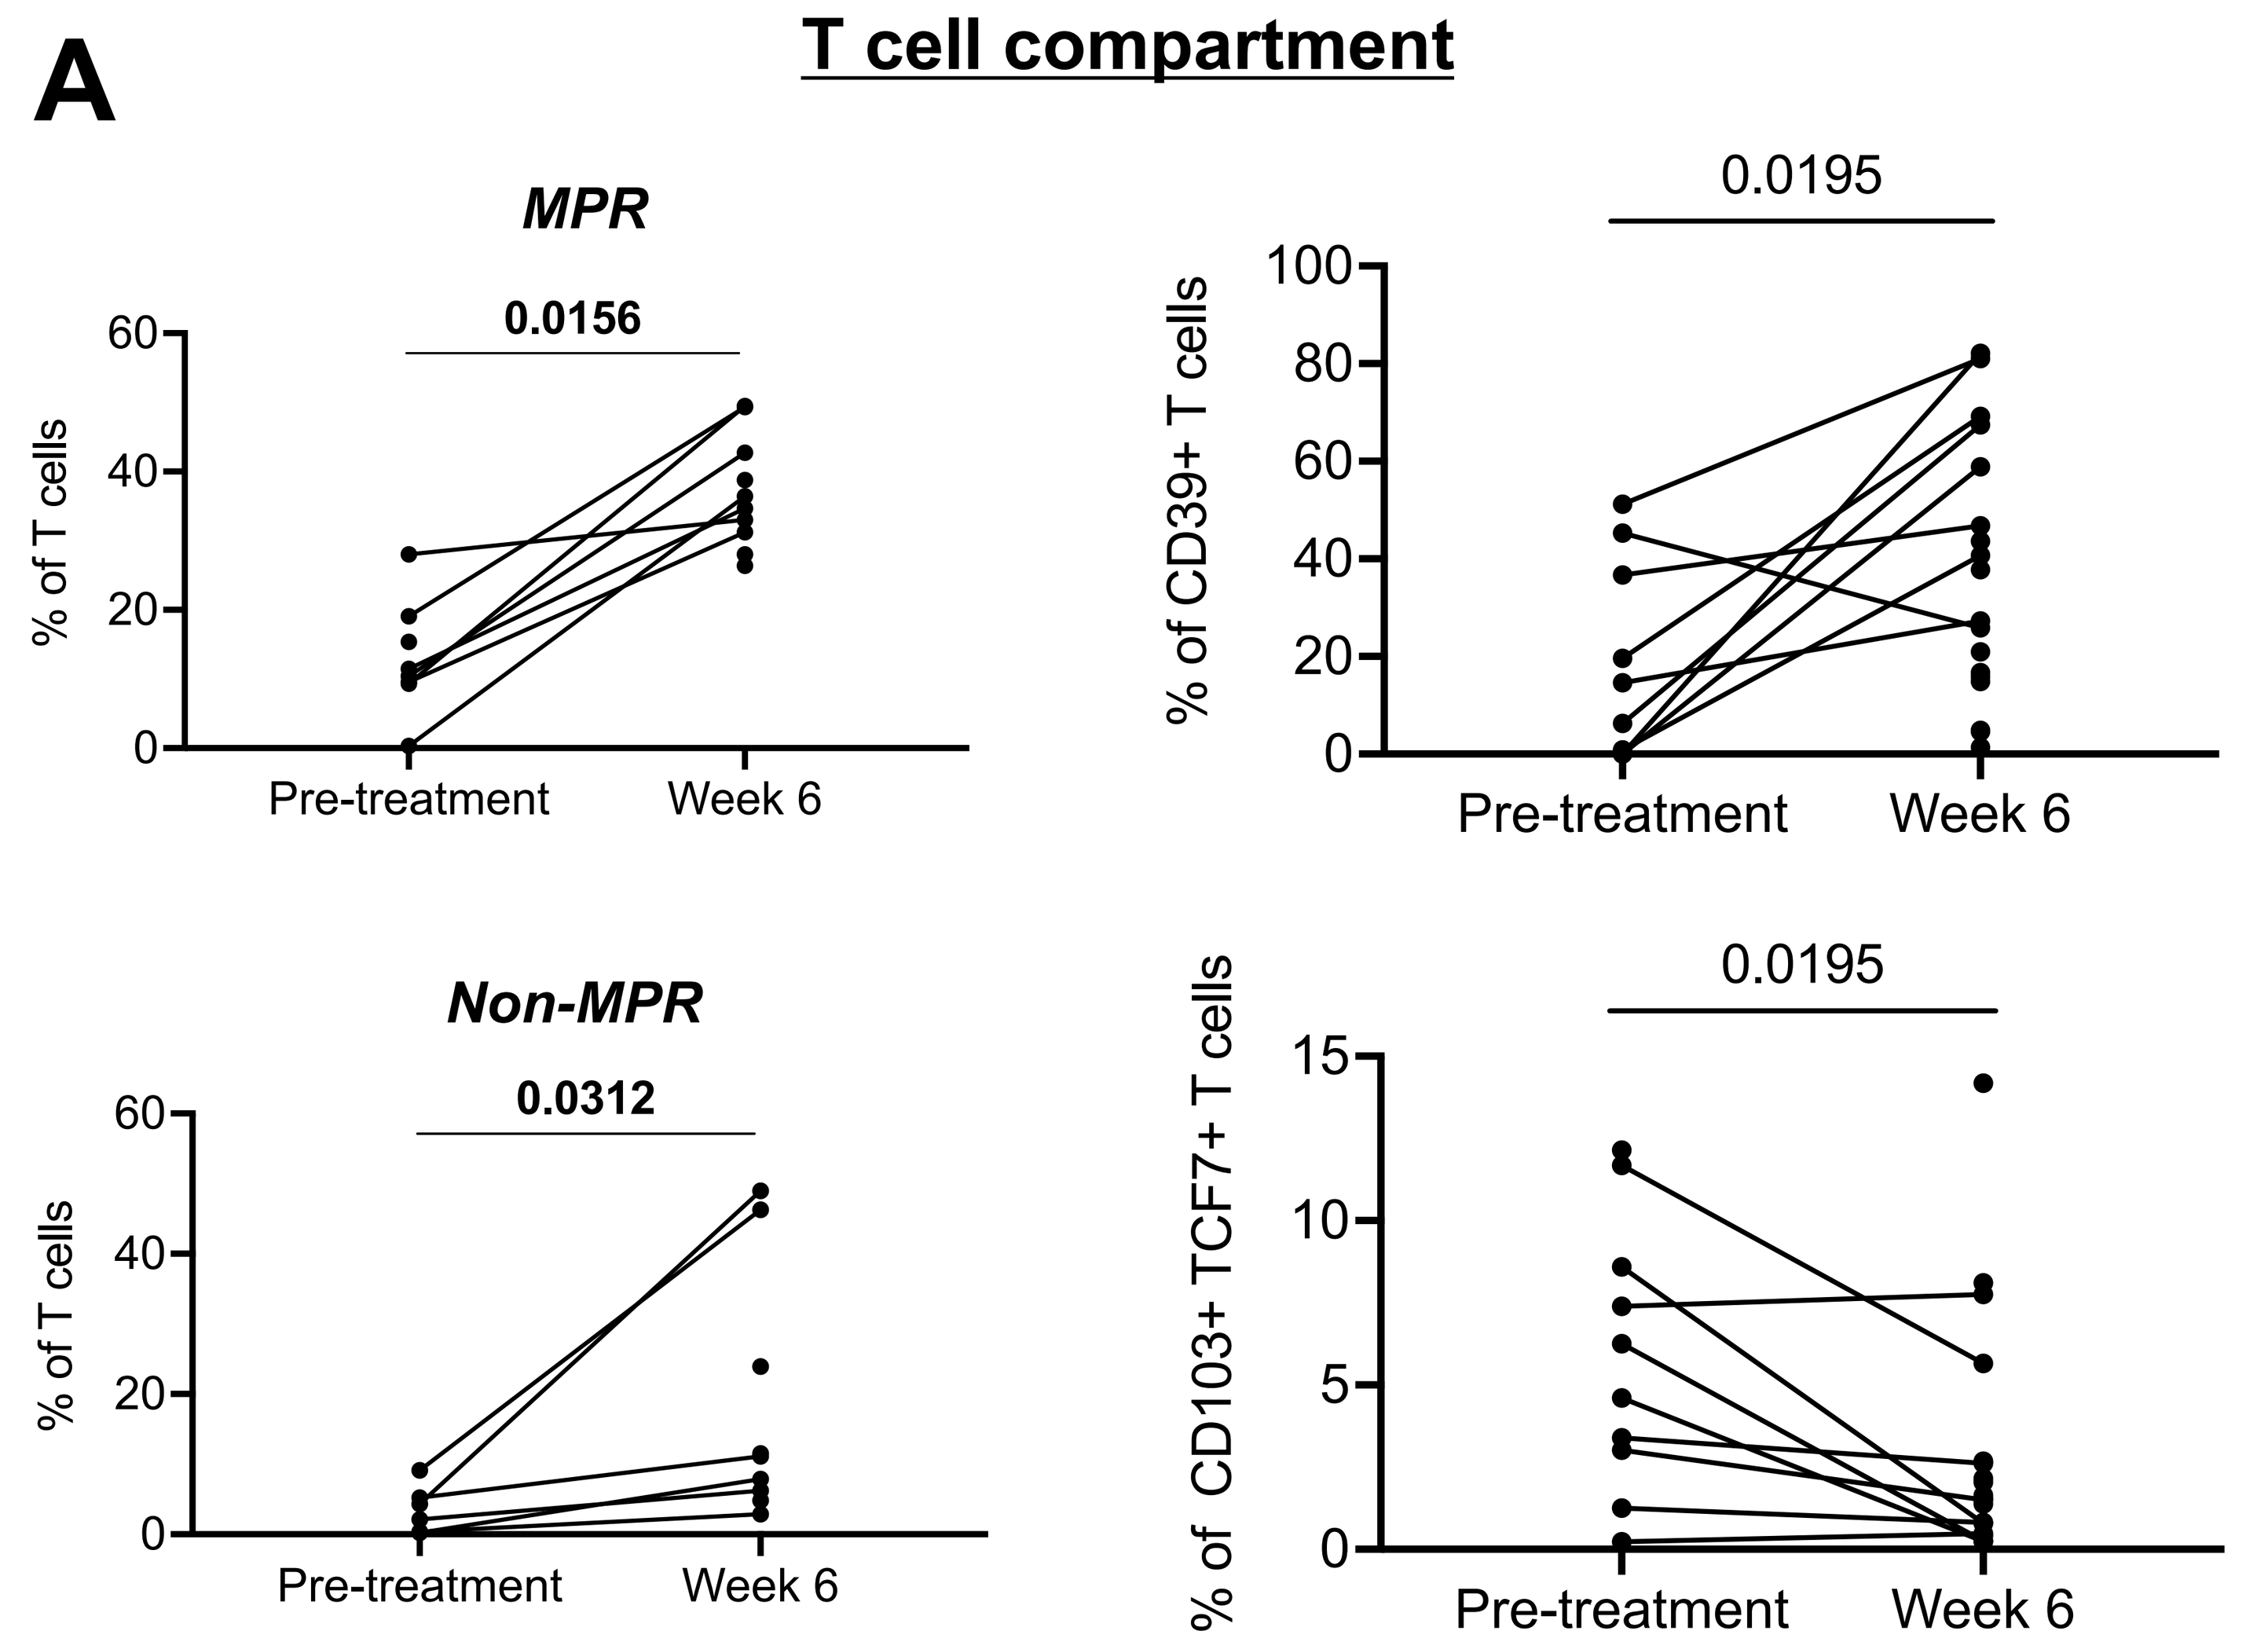
A
T cell compartment
MPR
0.0156
% of T cells
Non-MPR
0.0312
0.0195
% of CD39+ T cells
% of CD103+ TCF7+ T cells
Pre-treatment
Week 6

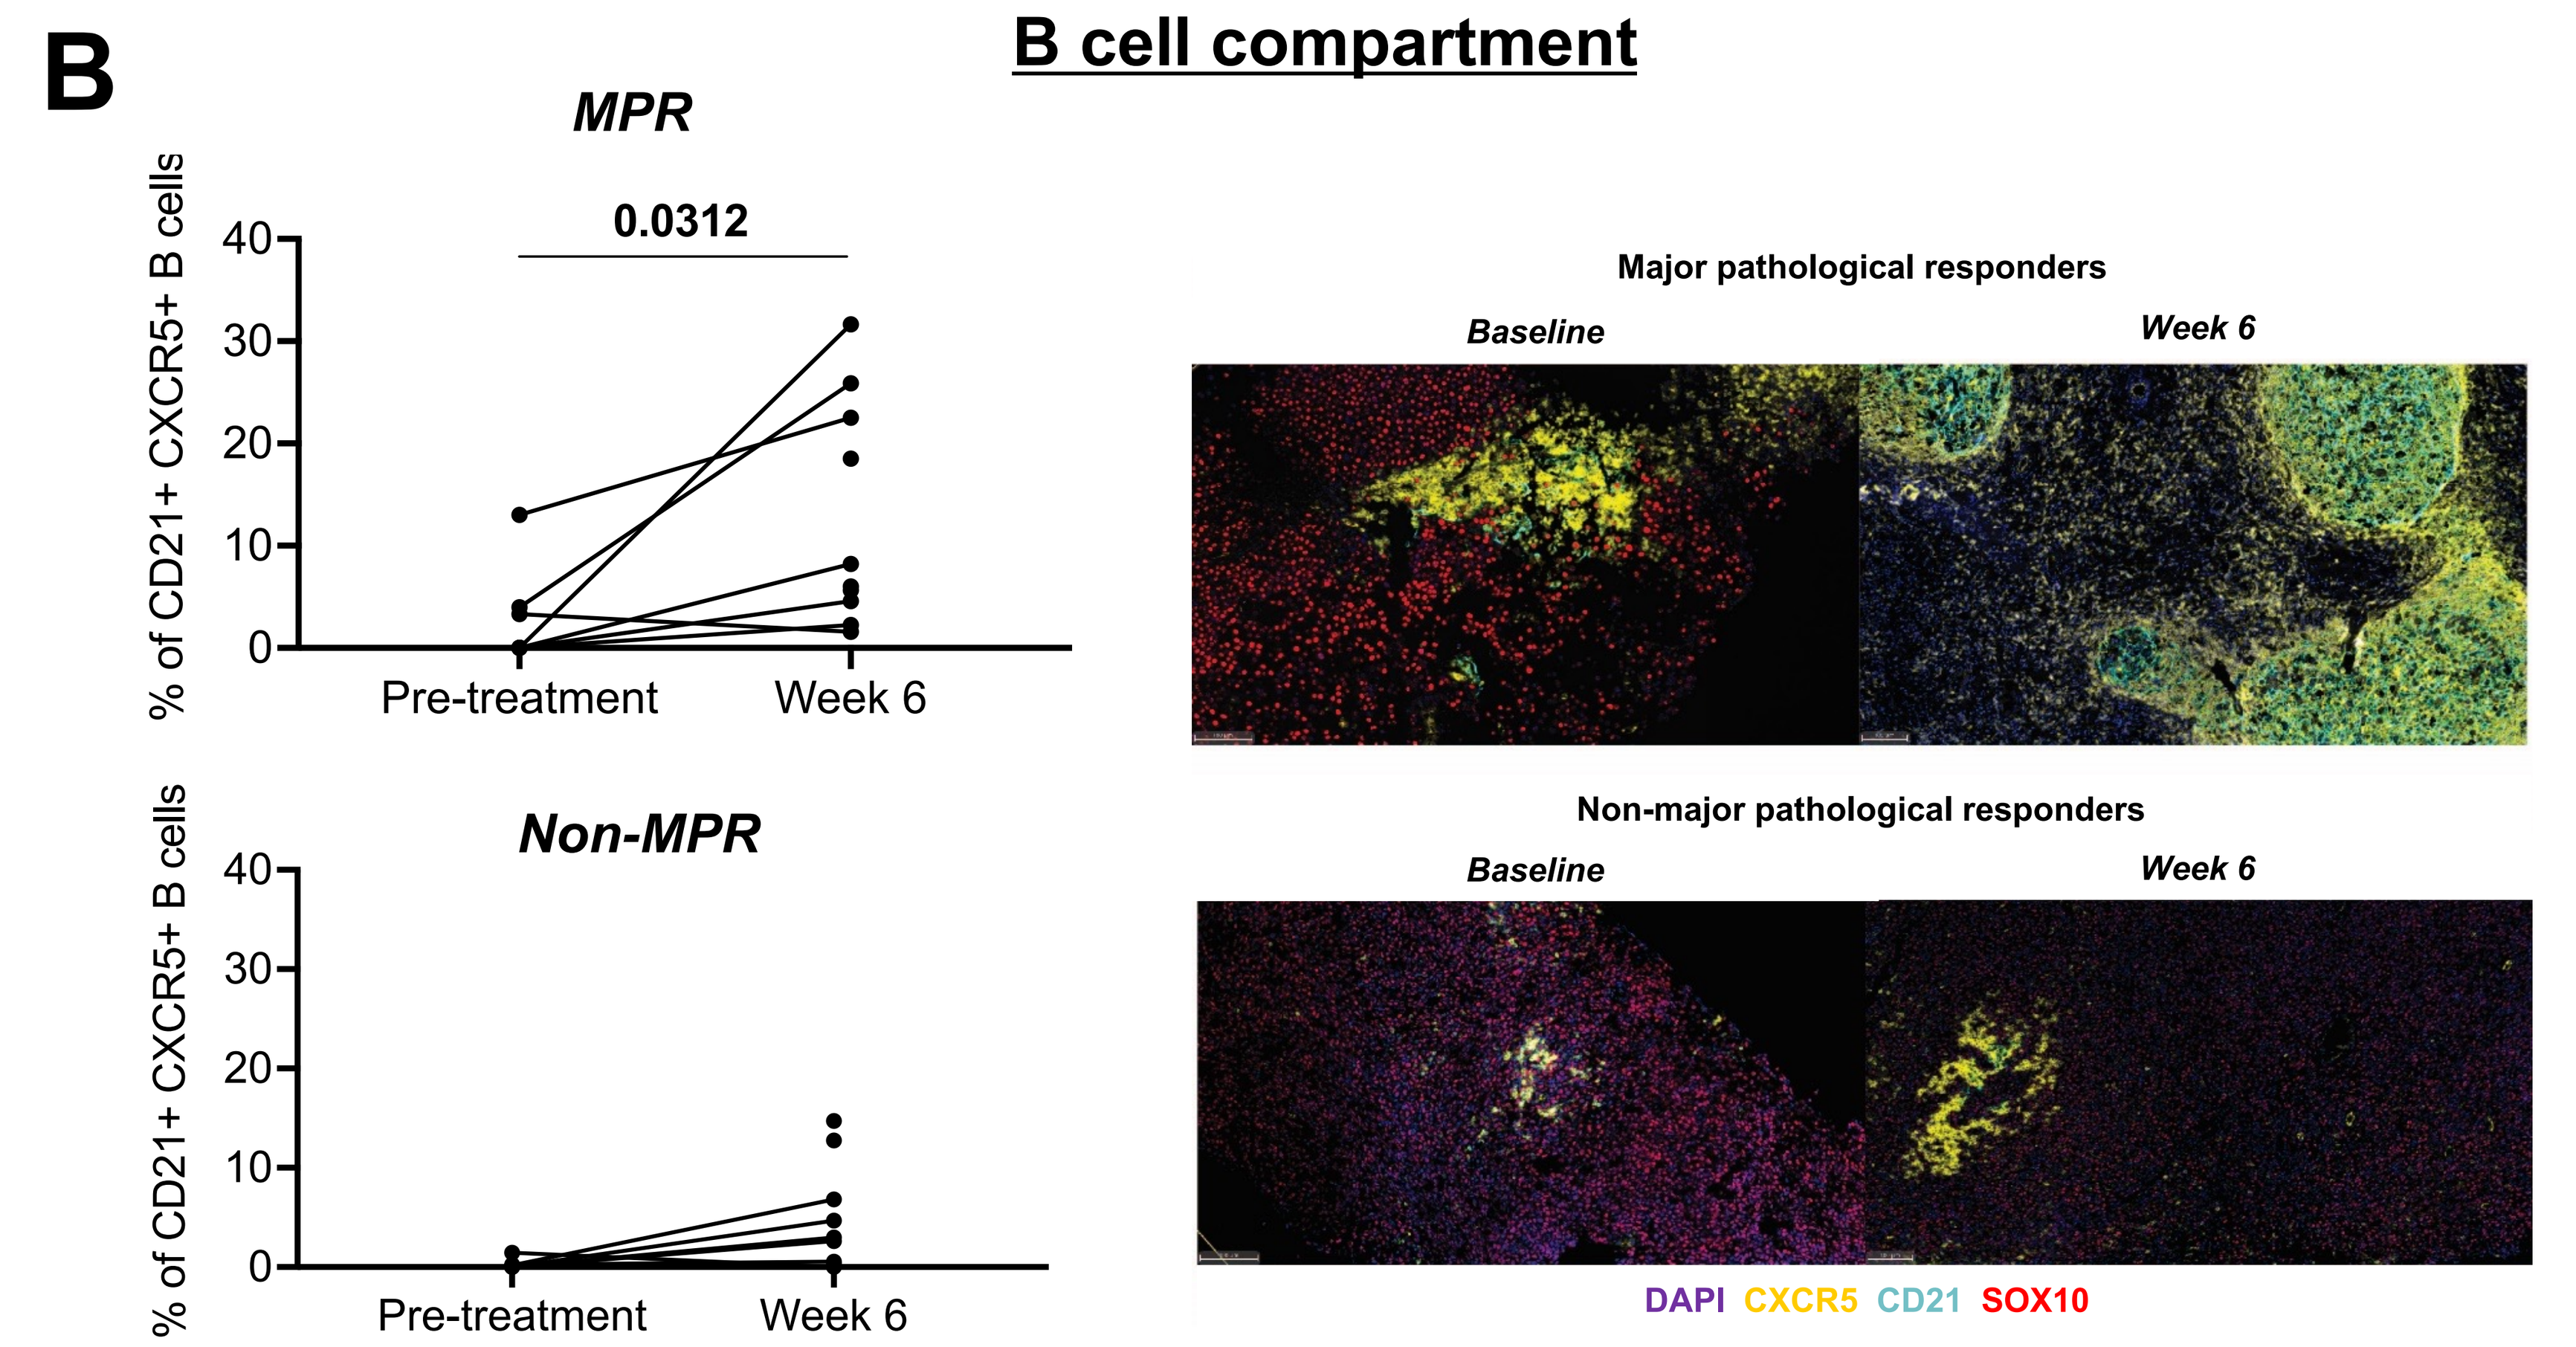
B
B cell compartment
MPR
0.0312
Non-MPR
% of CD21+ CXCR5+ B cells
Pre-treatment
Week 6
Major pathological responders
Non-major pathological responders
Baseline
Week 6
DAPI CXCR5 CD21 SOX10

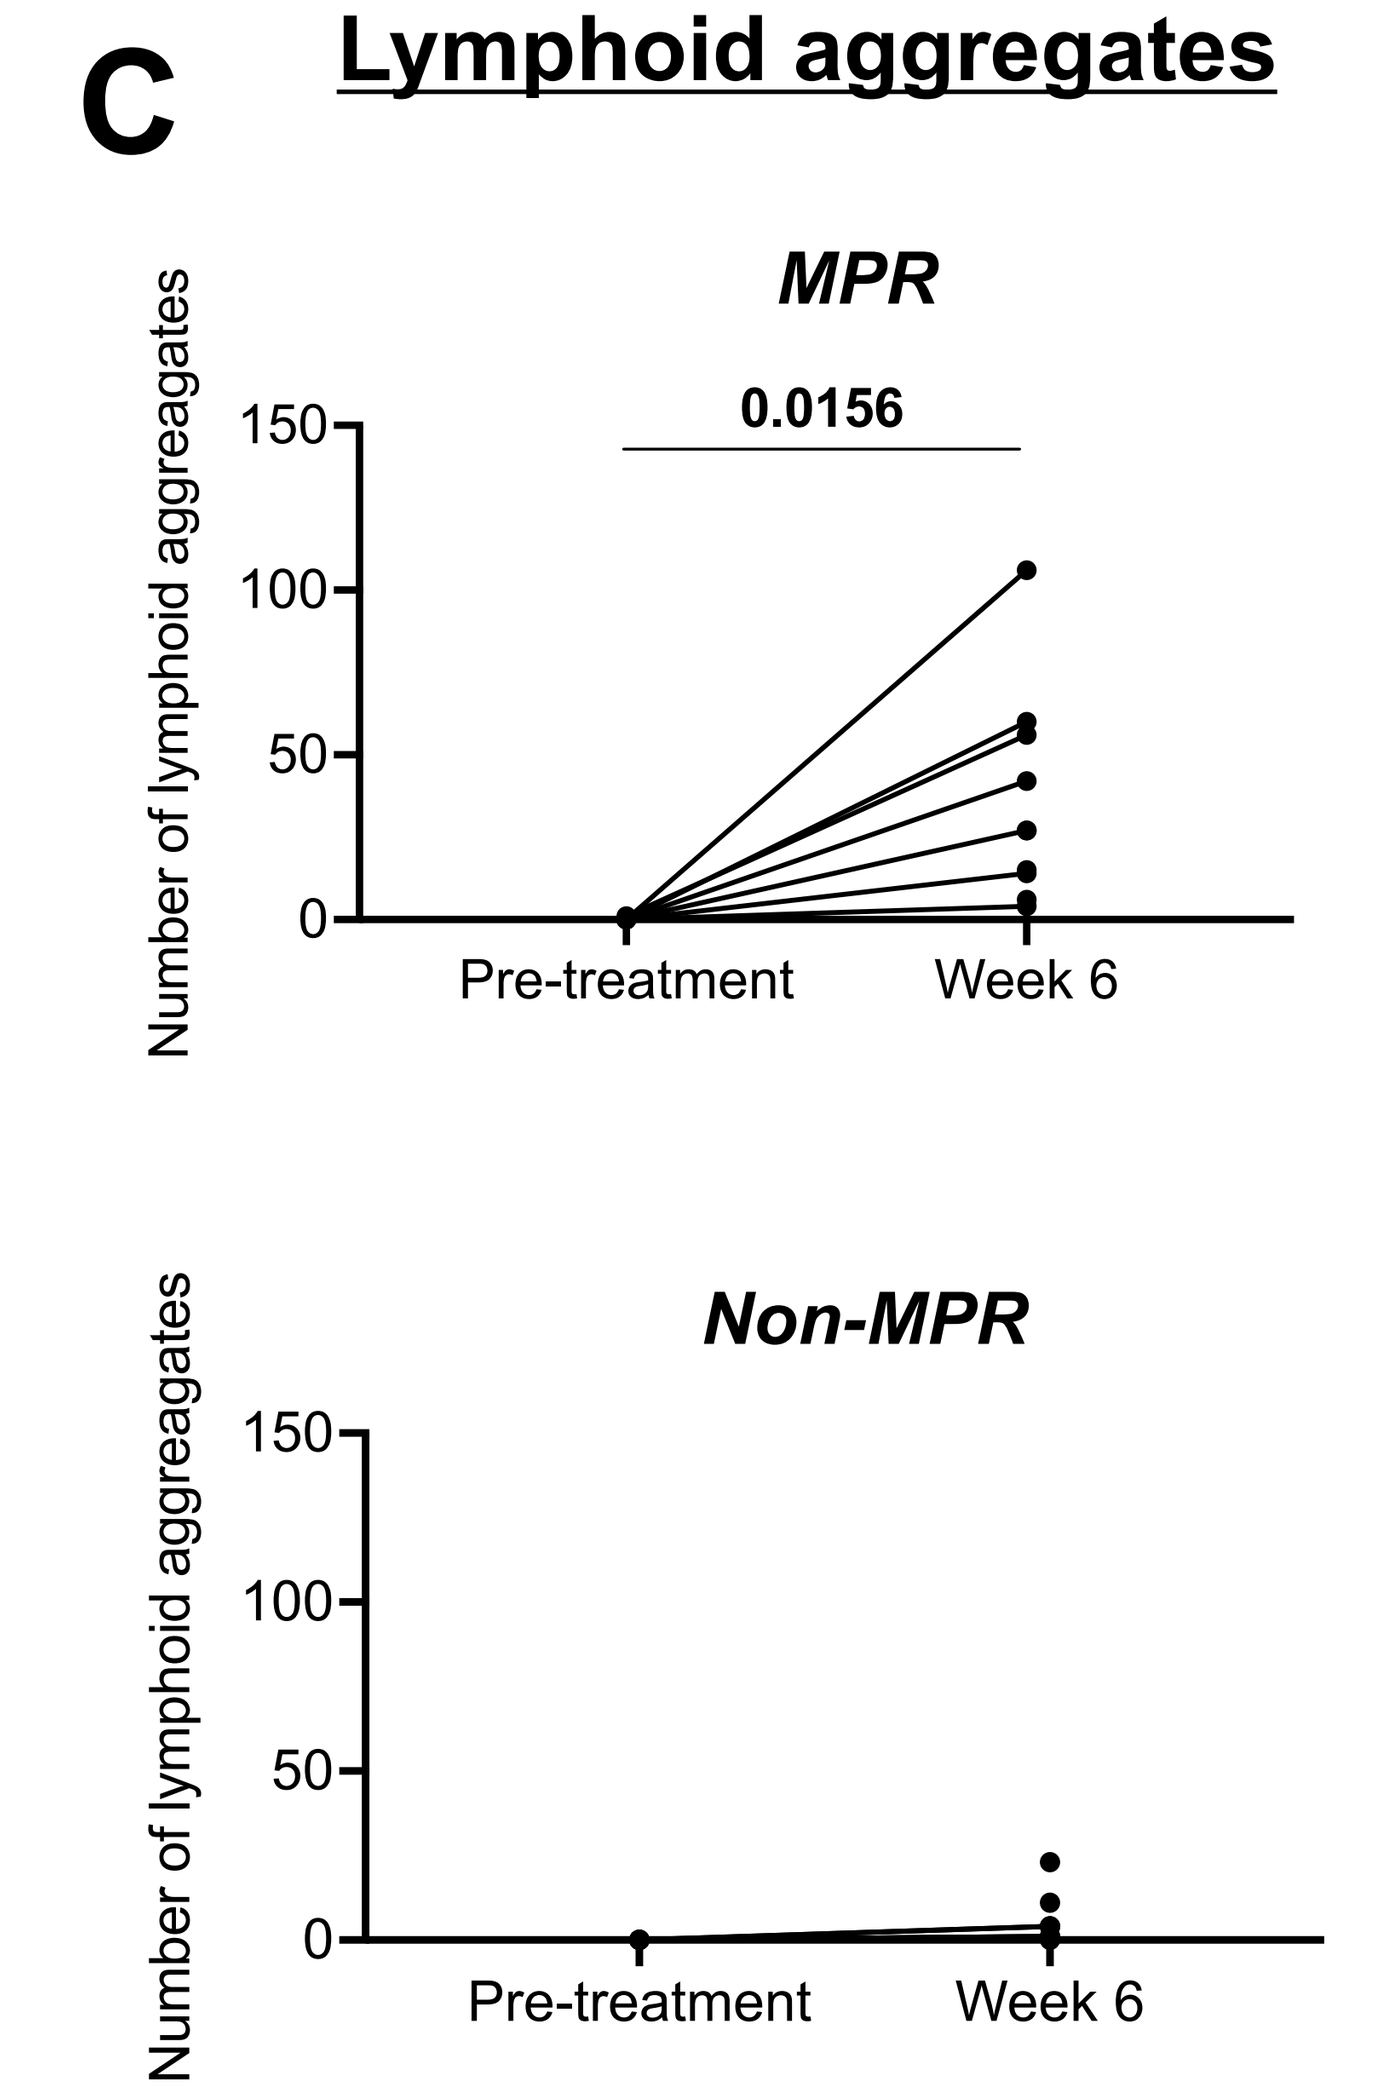
C
Lymphoid aggregates
MPR
0.0156
Non-MPR
Number of lymphoid aggregates
Pre-treatment
Week 6

FIGURE 3. Effect of neoIT on the TME in MPR *versus* non-MPR; analysis of the T cell (A) and B cell (B) compartments, as well as, of the lymphoid aggregates (C).

Conclusions

- ❑ ***MPR*** patients had a higher density of T cells in the TME at baseline, and a more differentiated and activated immune profile after neoIT compared with ***non-MPR*** patients. No difference was seen in the tdLNs between MPR *versus* Non-MPR.
- ❑ ***MPR*** patients had a significant increase in the number of lymphoid aggregates at week 6 compared to ***non-MPR*** patients; however, the role of these lymphoid aggregates, including the follicular B helper T cell subset and mature B cells, promoting a good pathological response to neoIT is yet to be clarified.

References

1. Patel SP, *et al*. NEJM 2023
2. Sade-Feldman M, *et al*. Cell 2018.
3. Siddiqui I, et al. Immunity 2019.
4. Long GV, et *al.* Presented at ESMO 2022; Abstract #3819

Acknowledgements

- All patients and their families
- Melanoma Institute Australia
- Lady Mary Fairfax Charitable Trust
- NeoPele trial supported by MSD (funding and study drug supply)

ASCO/2024 Poster #9566 Ines.silva@melanoma.org.au