FDG-PET Associations with Pathological Response and Survival with Neoadjuvant Immunotherapy for Melanoma

Li Zhou[1,2], Edward Hsiao[1,3], Serigne N. Lo[1,4], Matteo Carlino[1,5], George Au-Yeung[6,7], Maria Gonzalez[1], Andrew Spillane[1,3,4,8], Thomas E. Pennington[1,4,8,9], Julie Howle[1,4,5], Kerwin Shannon[1,4,10], Rony Kapoor[1,9], Jonathan Stretch[1,4,8,9], David E. Gyorki[6,7], Robyn P.M. Saw[1,4,8,9], Sydney Ch'ng[1,4,9], Richard A. Scolyer[1,4,9], Georgina V. Long[1,3,4,8], Alexander M. Menzies[1,3,4,8]

1. Melanoma Institute Australia; 2. Peking University Cancer Hospital; 3. Royal North Shore Hospital; 4. The University of Sydney; 5. Westmead Hospital; 6. Peter MacCallum Cancer Centre; 7. Sir Peter MacCallum Department of Oncology, University of Melbourne; 8. Mater Hospital; 9. Royal Prince Alfred Hospital; 10. Chris O'Brien Lifehouse

Melanoma Institute Australia

THE UNIVERSITY OF SYDNEY

Background

- Neoadjuvant anti-PD-1 monotherapy induces pathological response in 34% and pCR in 20% of patients with stage III melanoma, which is associated with patients' survival[1].
- Metabolic responses with ^{18}F-FDG PET/CT seen in metastatic melanoma associate with excellent long-term survival[2].
- ^{18}F-FDG-PET/CT imaging might be predictive for pathological response and recurrence-free survival in the neoadjuvant setting.

Objectives

- This study sought to explore associations between change in FDG-PET and either pathological response or recurrence-free survival (RFS).

Methods

Data from two prospective neoadjuvant clinical trial cohorts of stage III pts with RECIST measurable nodal melanoma were pooled and analysed:

1. NeoTrio[3] – 13 pts received 2 doses of pembrolizumab alone
2. NeoPele[4] – 20 pts received 2 doses of pembrolizumab and 5 weeks of lenvatinib

- All pts underwent baseline and week 6 (preoperative) FDG-PET assessments and all had surgery.
- PET responses were evaluated based on the modified EORTC criteria[5]. In addition to the standard categories: Complete Metabolic Response (CMR), Partial Metabolic Response (PMR), Stable Metabolic Disease (SMD), and Progressive Metabolic Disease (PMD), *a novel category was established, near-CMR, where the maximum standardized uptake value (SUVmax) decreased by more than 90% or residual uptake similar to background.*
- PET responses were determined while blinded to the outcome data. Pathological response was determined as per INMC criteria. Response was also assessed using RECIST criteria[6].

Results

Table 1. Correlations of FDG-PET and pathological response

FDG-PET response	Pathological Response: pCR	npCR	pPR	pNR	Total
CMR	1	1	0	0	2 (6%)
near CMR	5	1	0	0	6 (18%)
PMR	5	3	3	3	14 (42%)
SMD	0	0	2	3	5 (15%)
PMD	1	0	0	5	6 (18%)
Total	12 (36%)	5 (15%)	5 (15%)	11 (33%)	33

- 16 patients (48%) were BRAF V600E/K/R mutated.
- FDG-PET correlates closely with pathological response (concordant in green and discordant in yellow).

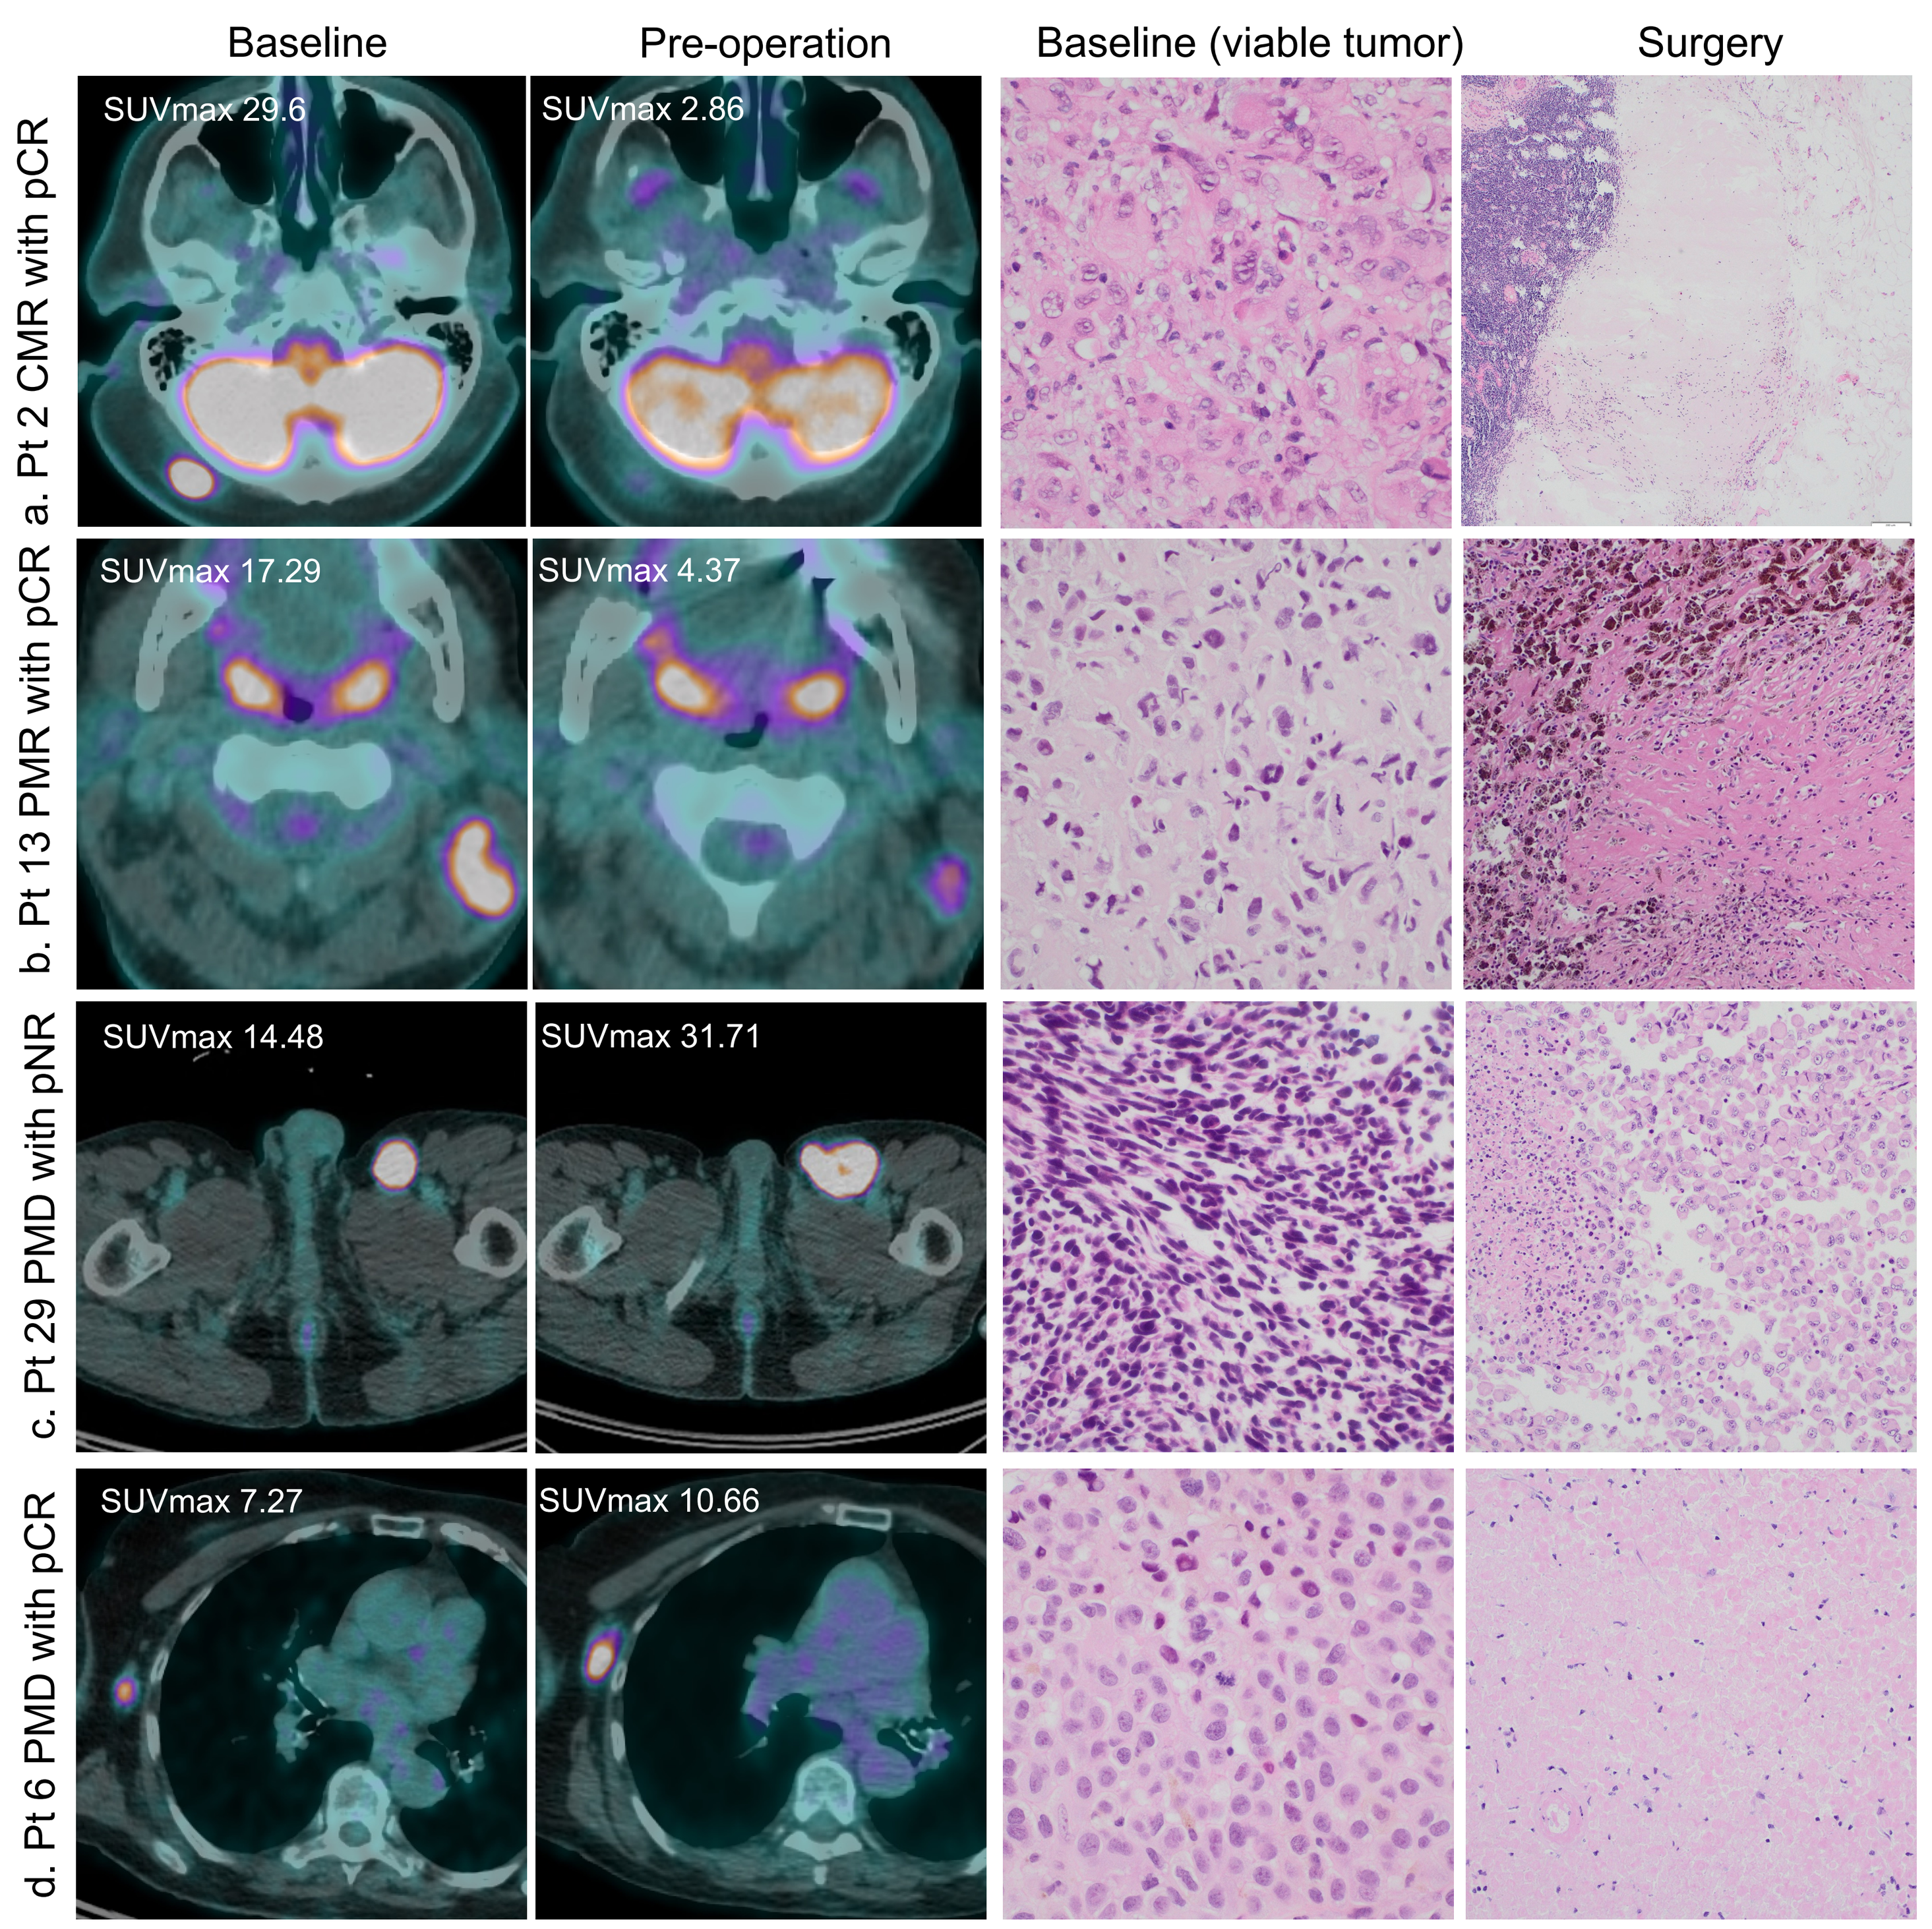
Baseline
Pre-operation
Baseline (viable tumor)
Surgery
a. Pt 2 CMR with pCR
b. Pt 13 PMR with pCR
c. Pt 29 PMD with pNR
d. Pt 6 PMD with pCR
SUVmax 29.6
SUVmax 2.86
SUVmax 17.29
SUVmax 4.37
SUVmax 14.48
SUVmax 31.71
SUVmax 7.27
SUVmax 10.66

Figure 1. Paired histology and radiographic images from 4 patients: a. Pt 2 with CMR received pCR (tumour bed fibrosis); b. Pt 13 with PMR received pCR (tumour bed with fibrosis and pigmented macrophages); c. Pt 29 with PMD and pNR (viable tumour with regions of necrosis); d. Pt 6 with PMD and pCR (necrotic tumour).

Figure 2. Proportion, RFS curves and COX models by pathological, metabolic and RECIST response categories

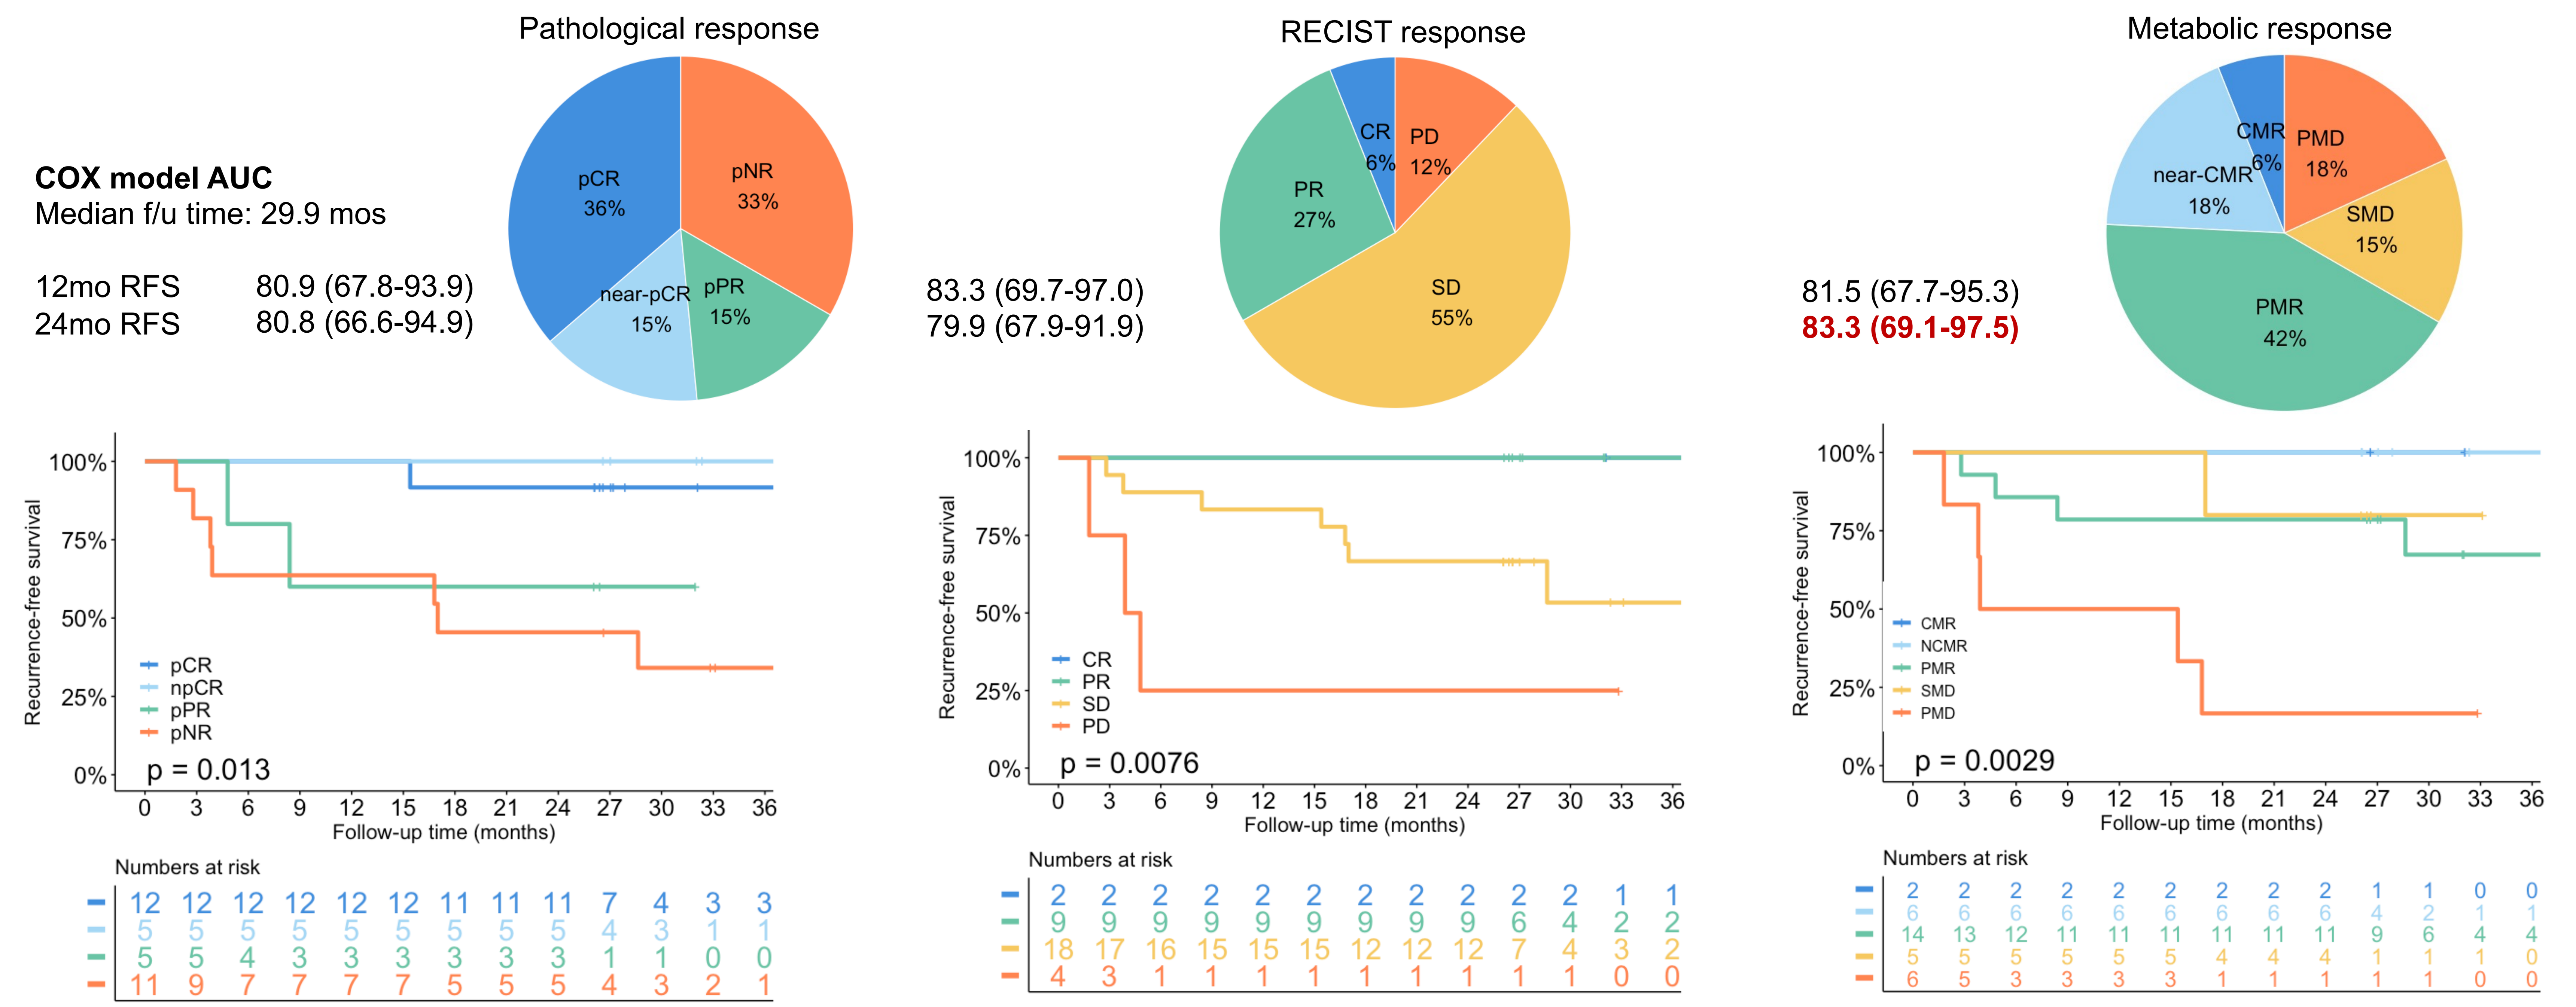
Pathological response
RECIST response
Metabolic response
COX model AUC
Median f/u time: 29.9 mos
12mo RFS 80.9 (67.8-93.9) 83.3 (69.7-97.0) 81.5 (67.7-95.3)
24mo RFS 80.8 (66.6-94.9) 79.9 (67.9-91.9) 83.3 (69.1-97.5)
pCR 36%
pNR 33%
near-pCR 15%
pPR 15%
CR 6%
PD 12%
PR 27%
SD 55%
CMR 6%
PMD 18%
near-CMR 18%
SMD 15%
PMR 42%
p = 0.013
p = 0.0076
p = 0.0029
Recurrence-free survival
Follow-up time (months)
Numbers at risk

- FDG-PET is more accurate than CT and pathological response in predicting 24mo-RFS with neoadjuvant immunotherapy in melanoma.

Figure 3. Waterfall plot, box plot and RFS curves of SUVmax change association with pathological response

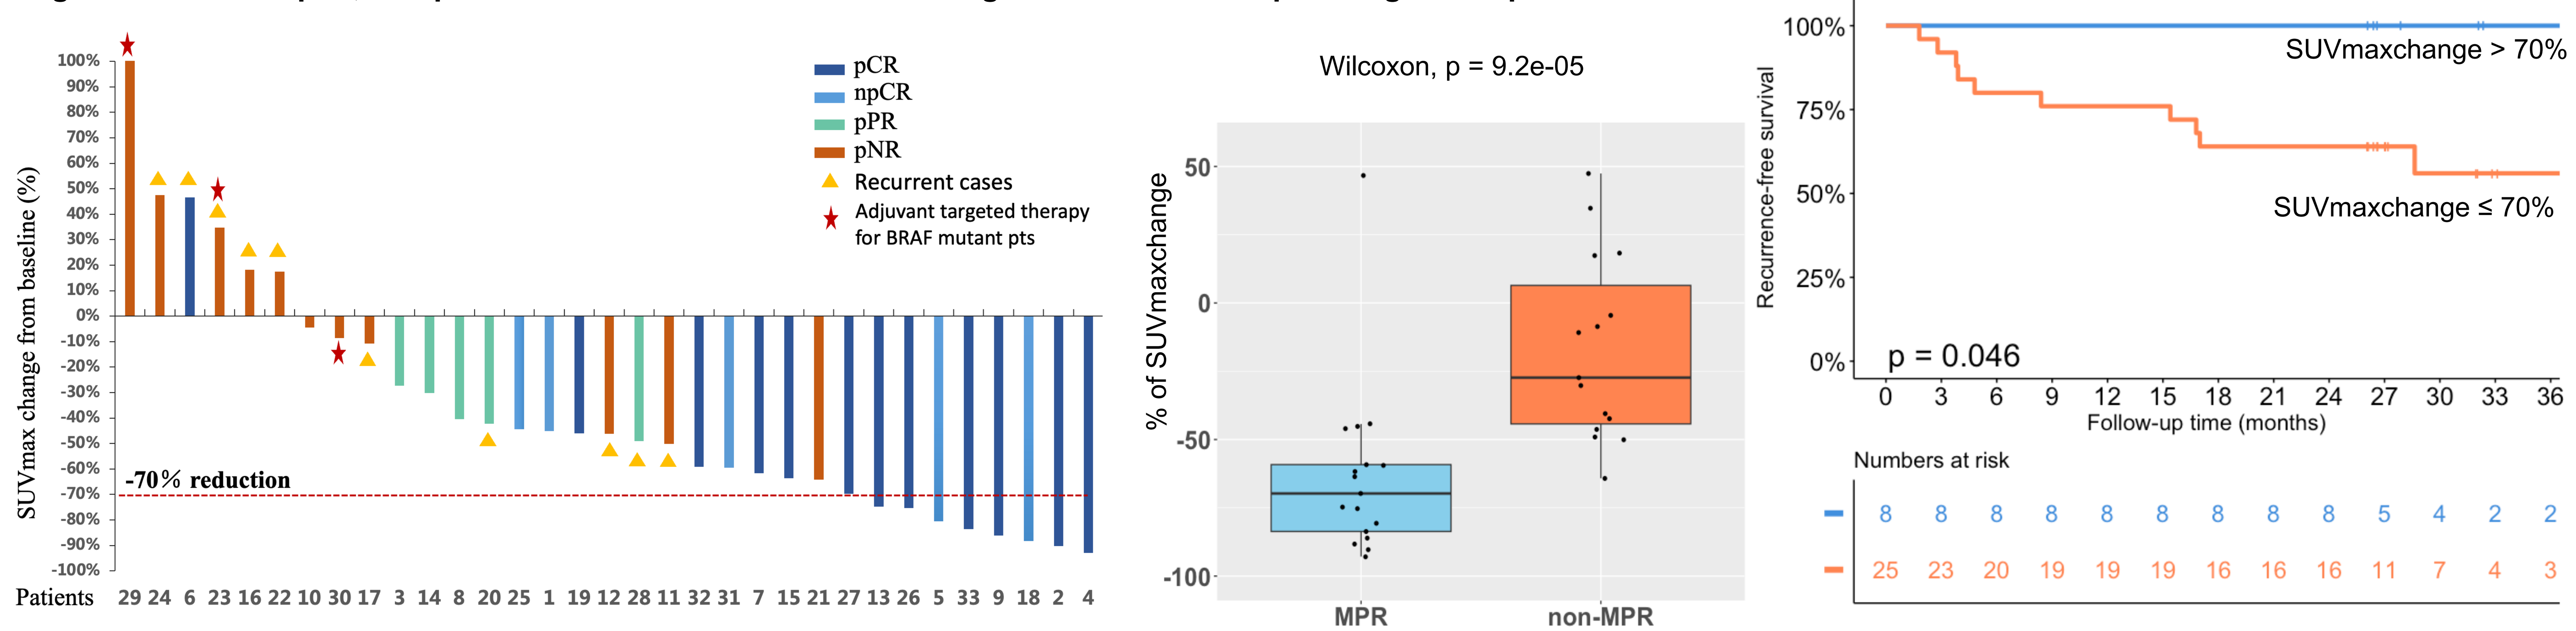
pCR
npCR
pPR
pNR
Recurrent cases
Adjuvant targeted therapy for BRAF mutant pts
-70% reduction
SUVmax change from baseline (%)
Patients
Wilcoxon, p = 9.2e-05
% of SUVmaxchange
MPR
non-MPR
SUVmaxchange > 70%
SUVmaxchange ≤ 70%
p = 0.046
Recurrence-free survival
Follow-up time (months)
Numbers at risk

- The extent of SUVmax decrease correlates with pathological response and RFS.

Conclusions

- FDG-PET demonstrates good utility in predicting pathological response and survival with neoadjuvant immunotherapy in melanoma.
- PET may identify pts who are going to be pathological responders and who may de-escalate surgery, or non-responders who may benefit from an alternative neoadjuvant therapy prior to surgery.

References

1. Menzies A, et al. Nature Medicine, 2021.
2. Dimitriou F, et al. Annals of Oncology, 2021.
3. Long G, et al. NeoTrio; ASCO 2022.
4. Long G, et al. NeoPeLe; ESMO 2022.
5. Young H, et al. Eur J Cancer, 1999.
6. Eisenhauer EA, et al. Eur J Cancer, 2009.

Acknowledgements

- All patients and their families
- Trial sponsored by the Melanoma Institute Australia
- Drug and funding support by Merck Sharp & Dohme (Australia) Pty Ltd and Novartis

ASCO/2024 Poster #9523
alexander.menzies@sydney.edu.au